Developing a potent gene therapy candidate for facioscapulohumeral muscular dystrophy (FSHD) through high throughput AAV capsid and cargo engineering

Saurav Seshadri, Edward Marsh, Kyle Brown, Shayan Tabebordbar, Jana Jenquin, Janelle Stricker, Brian Ferguson, Mark Fielden, Sharif Tabebordbar
Kate Therapeutics, San Diego, CA

Background

- Facioscapulohumeral muscular dystrophy (FSHD) is an autosomal-dominant genetic disorder affecting an estimated 1 in 8,000 individuals.
- Patients with FSHD typically exhibit progressive wasting of muscles in the face, shoulders, upper arms, legs, and abdomen; approximately 20% of patients will eventually require the use of a wheelchair, and the majority experience debilitating pain and fatigue.
- De-repression of the transcription factor *DUX4* has been identified as the pathogenetic mechanism in FSHD: insufficient epigenetic silencing of the *DUX4* gene in patient muscles results in aberrant expression of DUX4, which is toxic to mature myofibers.
- KateTx is pursuing knockdown of *DUX4* transcript using gene therapy as a therapeutic strategy for FSHD.

Therapeutic Approach

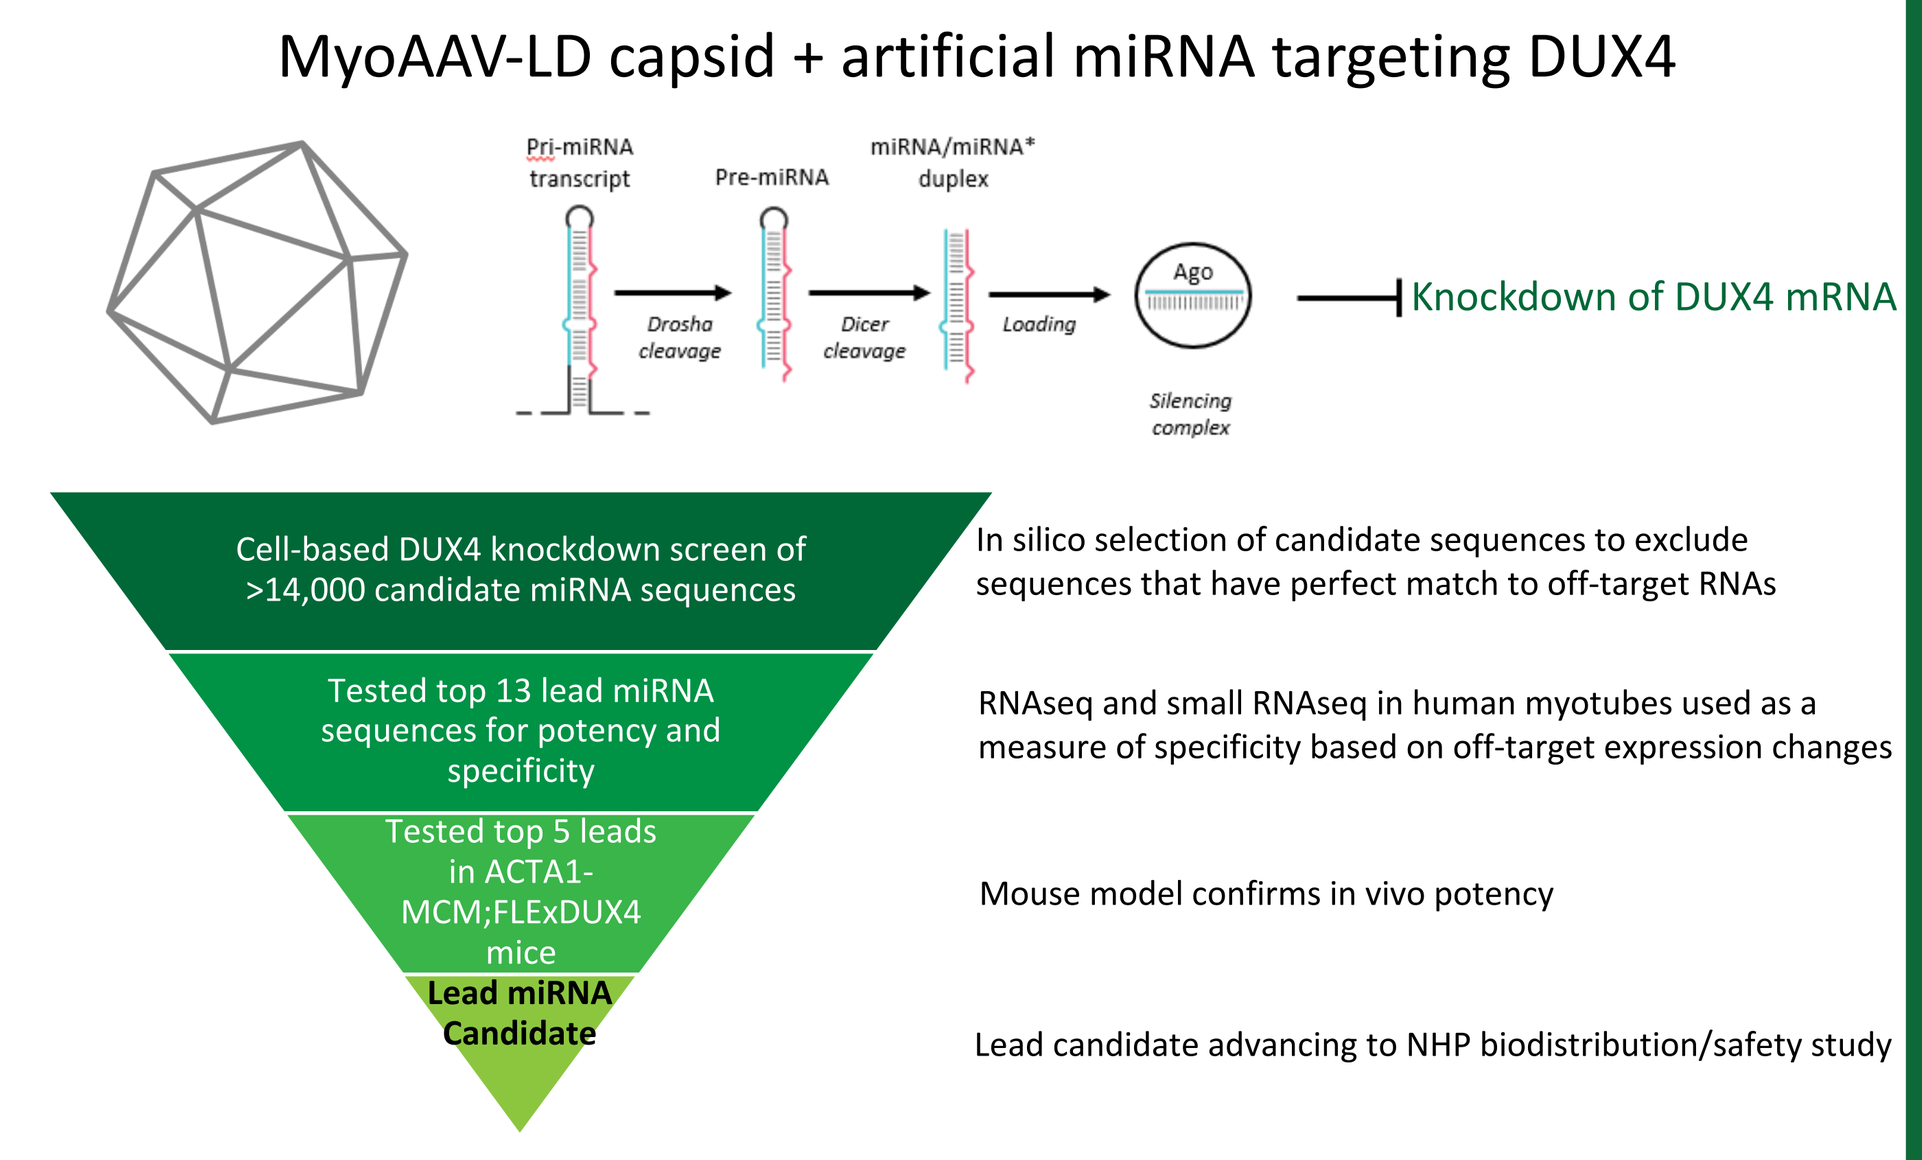
MyoAAV-LD capsid + artificial miRNA targeting DUX4
Pri-miRNA transcript
Pre-miRNA
miRNA/miRNA* duplex
Drosha cleavage
Dicer cleavage
Loading
Ago
Silencing complex
Knockdown of DUX4 mRNA
Cell-based DUX4 knockdown screen of >14,000 candidate miRNA sequences
Tested top 13 lead miRNA sequences for potency and specificity
Tested top 5 leads in ACTA1-MCM;FLExDUX4 mice
Lead miRNA Candidate
In silico selection of candidate sequences to exclude sequences that have perfect match to off-target RNAs
RNAseq and small RNAseq in human myotubes used as a measure of specificity based on off-target expression changes
Mouse model confirms in vivo potency
Lead candidate advancing to NHP biodistribution/safety study

References

1. https://www.fshdsociety.org/what-is-fshd/
2. Tabebordbar et al, 2021 https://doi.org/10.1016/j.cell.2021.08.028
3. Wang & Tawil, 2021 https://content.iospress.com/articles/journal-of-neuromuscular-diseases/jnd200554
4. Wallace et al, 2012 10.1038/mt.2012.68
5. Krom et al, 2012 10.1016/j.ajpath.2012.07.007
6. Jones et al, 2020 https://doi.org/10.1186/s13395-020-00227-4

Tiling screen identifies the most potent artificial miRNA sequences targeting DUX4

- Generated a HEK293 cell line stably expressing mCherry-tagged DUX4 (Hox1 mutant).
- Performed a lentiviral/FACS-based screen to identify the most potent artificial miRNA sequences targeting DUX4.
- Selected several potent sequences for individual testing in FSHD patient-derived myotubes.

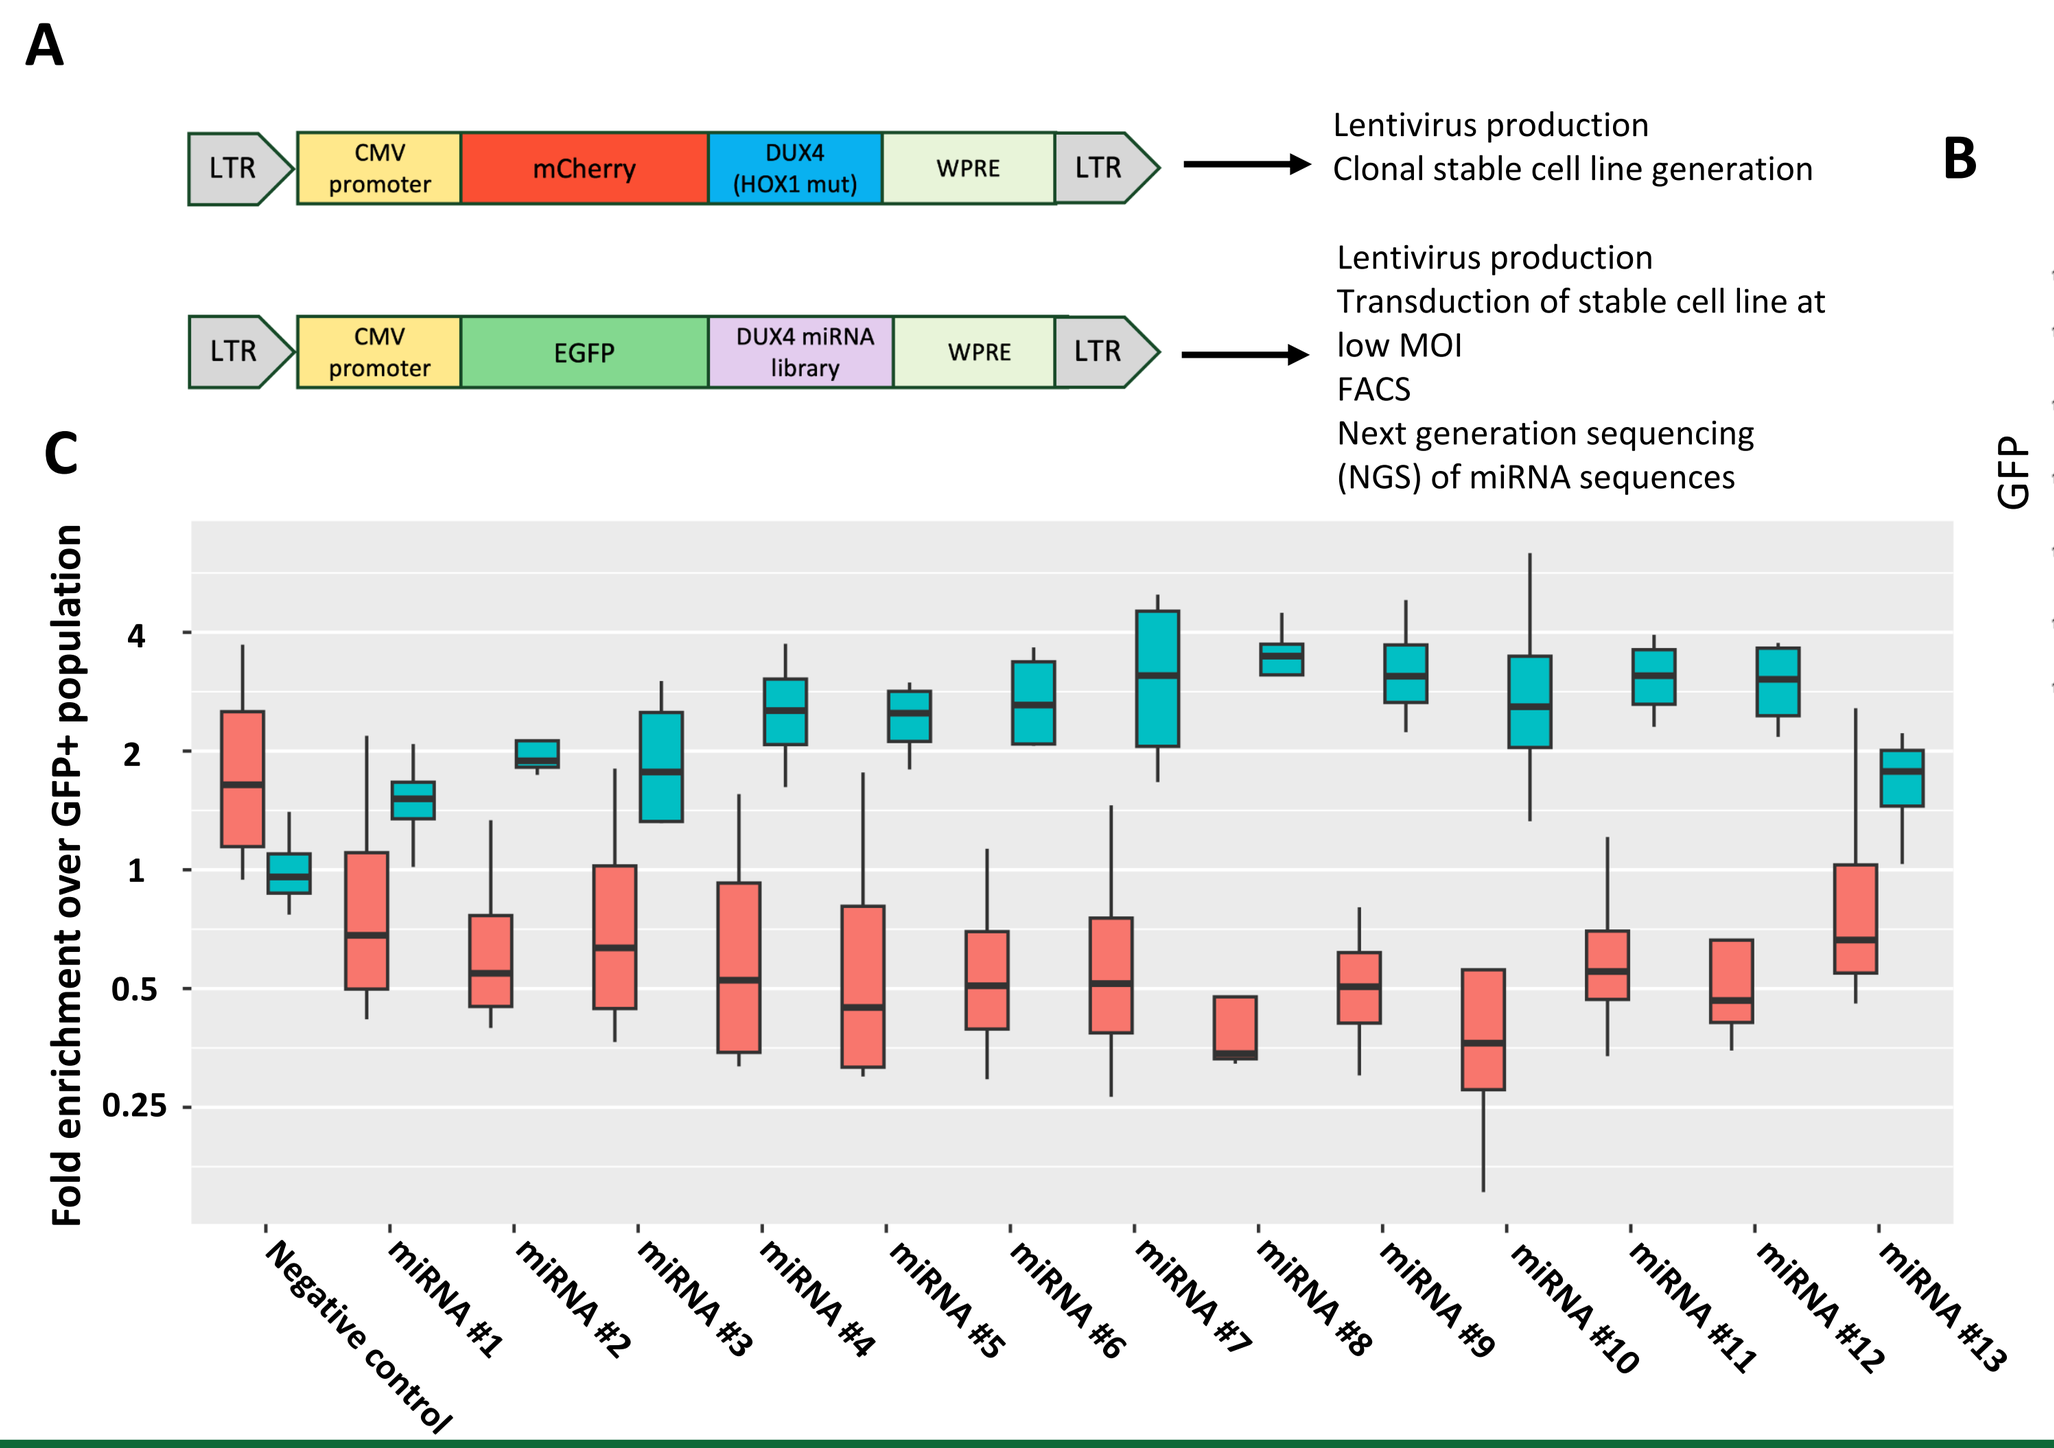
A
LTR
CMV promoter
mCherry
DUX4 (HOX1 mut)
WPRE
LTR
Lentivirus production
Clonal stable cell line generation
EGFP
DUX4 miRNA library
Lentivirus production
Transduction of stable cell line at low MOI
FACS
Next generation sequencing (NGS) of miRNA sequences
C
Fold enrichment over GFP+ population
Negative control
miRNA #1
miRNA #2
miRNA #3
miRNA #4
miRNA #5
miRNA #6
miRNA #7
miRNA #8
miRNA #9
miRNA #10
miRNA #11
miRNA #12
miRNA #13

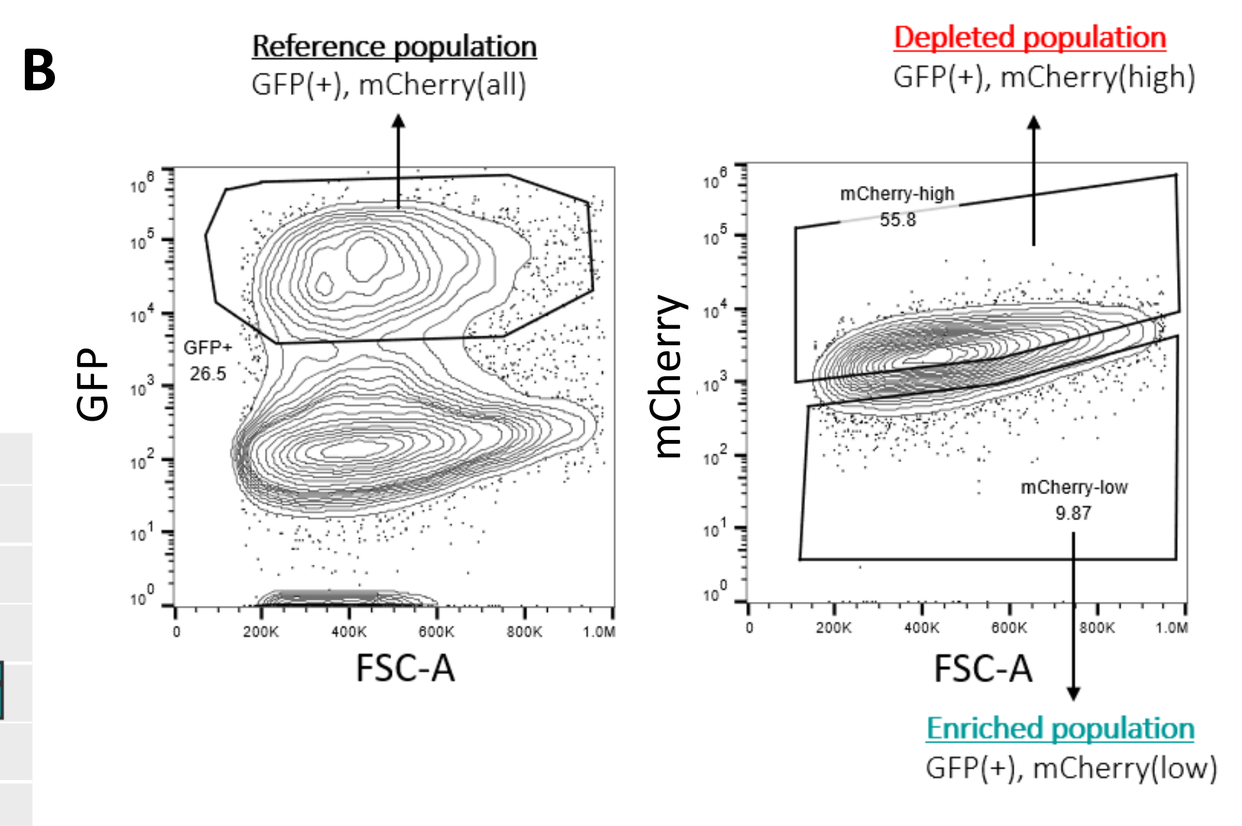
B
Reference population
GFP(+), mCherry(all)
Depleted population
GFP(+), mCherry(high)
GFP
mCherry
FSC-A
mCherry-high 55.8
mCherry-low 9.87
Enriched population
GFP(+), mCherry(low)

Figure 1: Tiling screening of the DUX4 transcript for effective artificial miRNAs identified several hits that enable potent DUX4 knockdown.
A) Schematic of lentiviral constructs used in the screen
B) FACS workflow and selection of positive and negative populations
C) List of top hit miRNA sequences based on their enrichment in the GFP+ mCherry (low) population

KateTx FSHD lead candidate effectively knocks down DUX4 in FSHD patient myotubes

- Top miRNA hits from the screen knock down DUX4 when tested individually in FSHD1 patient myotubes.
- KateTx FSHD lead candidate potently knocks down DUX4 and decreases expression of DUX4 downstream genes, regardless of the timing of transduction.

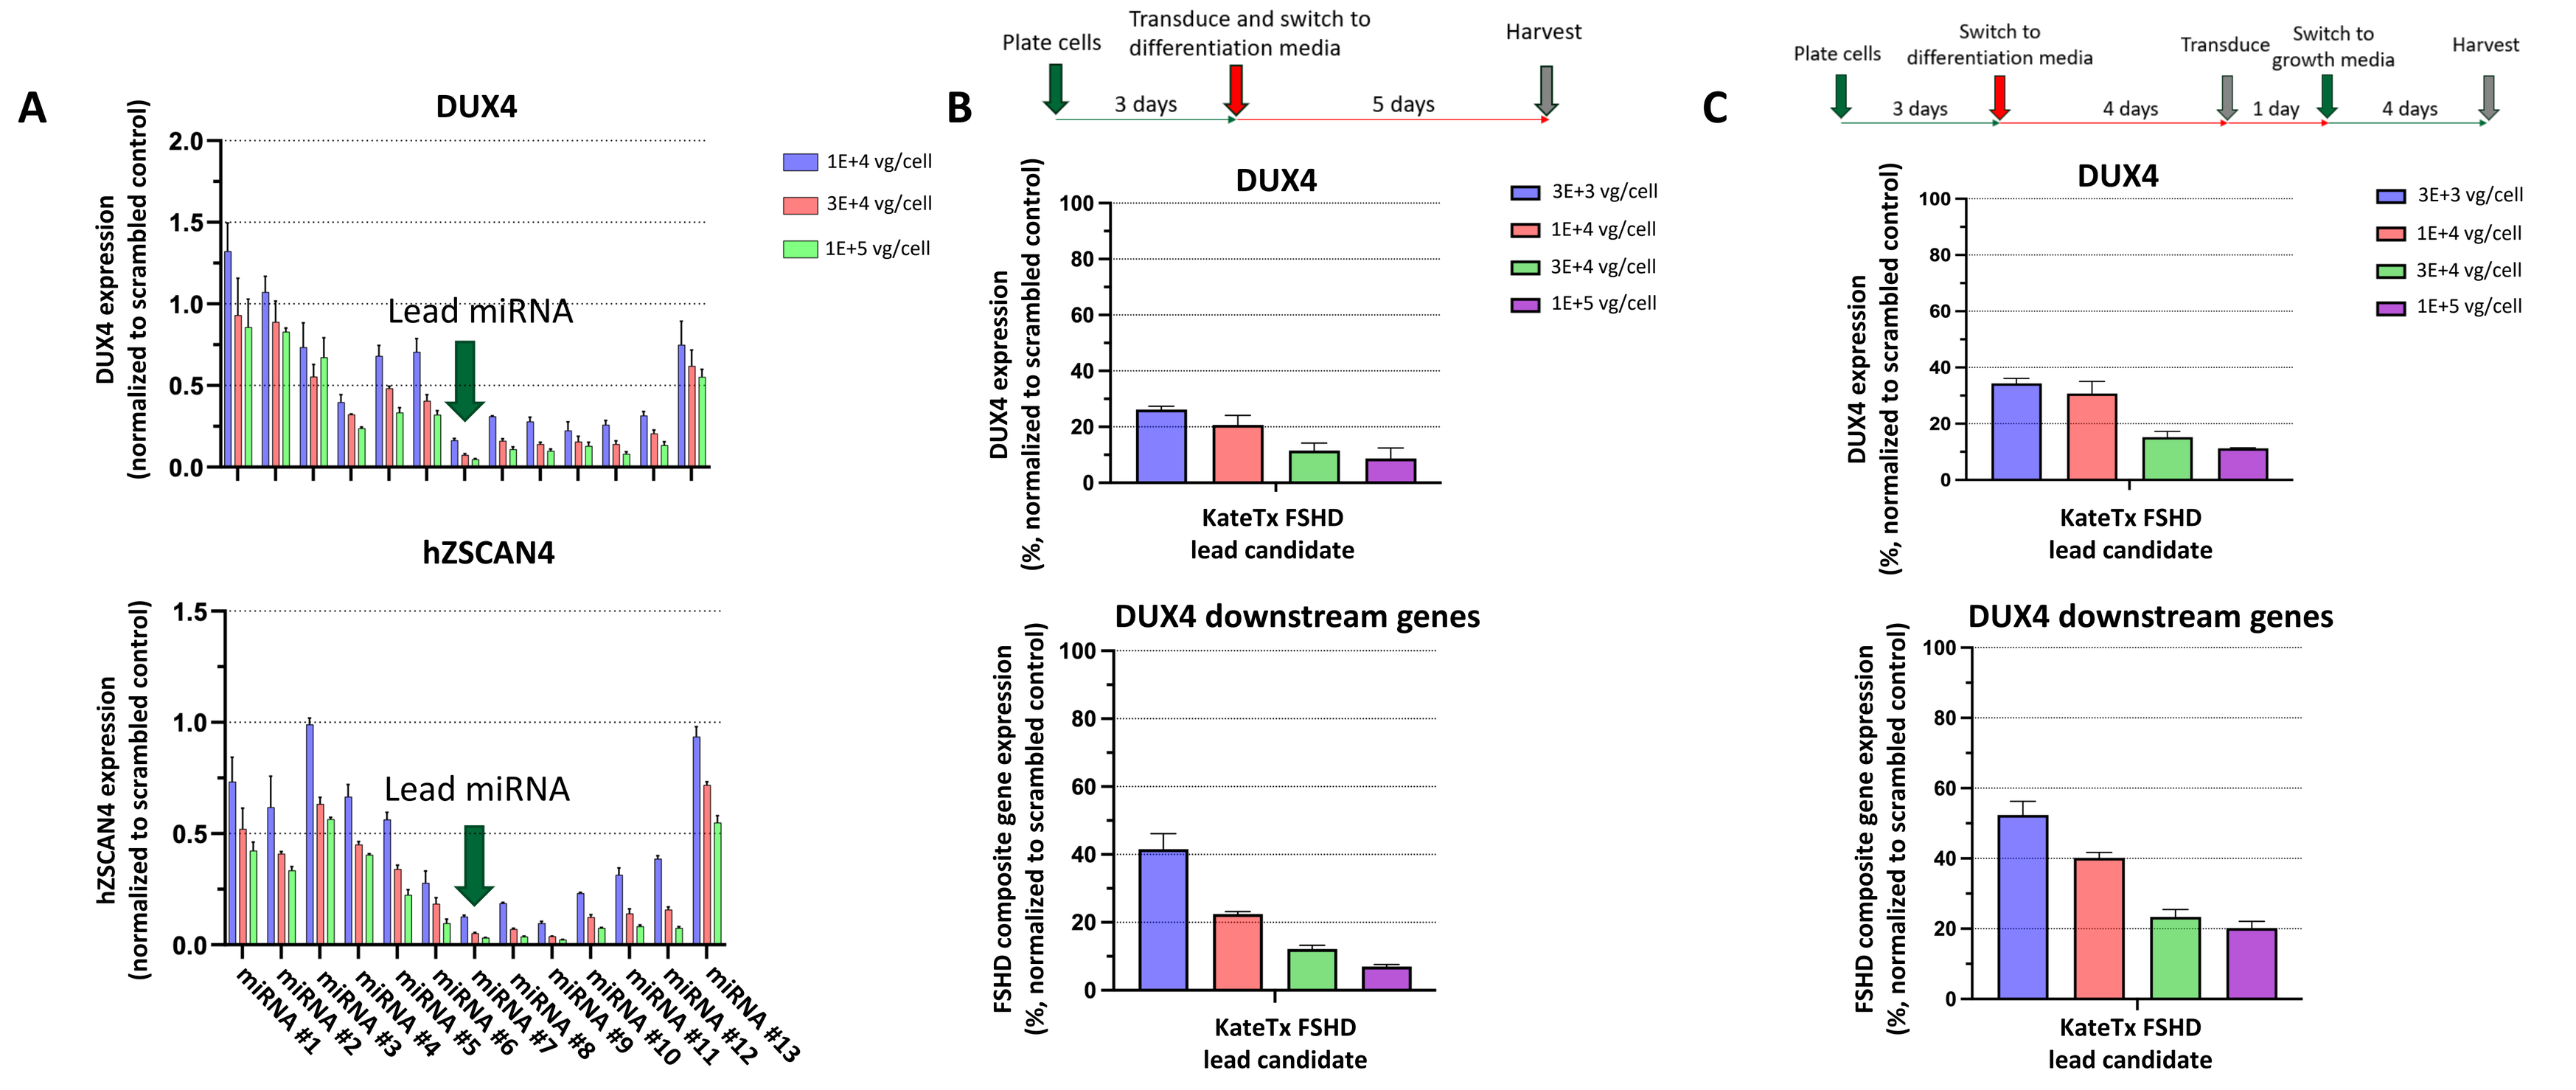
A
DUX4
hZSCAN4
Lead miRNA
1E+4 vg/cell
3E+4 vg/cell
1E+5 vg/cell
B
Plate cells
Transduce and switch to differentiation media
Harvest
3 days
5 days
C
Switch to differentiation media
Transduce
Switch to growth media
4 days
1 day
DUX4 downstream genes
KateTx FSHD lead candidate
3E+3 vg/cell

Figure 2: KateTx FSHD lead candidate effectively knocks down DUX4 and decreases expression of DUX4 downstream genes in FSHD patient cells.
A) Expression of DUX4 and DUX4 downstream gene hZSCAN4 in FSHD patient myotubes transduced with top hits from tiling screen
B) KateTx FSHD lead candidate knocks down DUX4 in FSHD patient myotubes in a dose dependent manner when cells are transduced at the time of inducing differentiation. FSHD composite gene expression was calculated by averaging normalized expression of several DUX4 downstream genes (CCNA1, MBD3L2, ZSCAN4, TRIM43, KHDC1L).
C) KateTx FSHD lead candidate knocks down DUX4 in FSHD patient myotubes in a dose dependent manner when myotubes are transduced after differentiation.

KateTx FSHD lead candidate does not result in off-target effects based on RNAseq and small RNAseq

- Transduced differentiated human primary myotubes with the KateTx FSHD lead candidate or a scrambled control.
- Confirmed that lead miRNA was expressed at therapeutic levels.
- Performed RNAseq and small RNAseq to assess differential gene and endogenous miRNA expression between myotubes transduced with the lead candidate and scrambled control. No endogenous mRNA or miRNAs met criteria of adjusted p<0.001, 33% decrease or 50% increase.

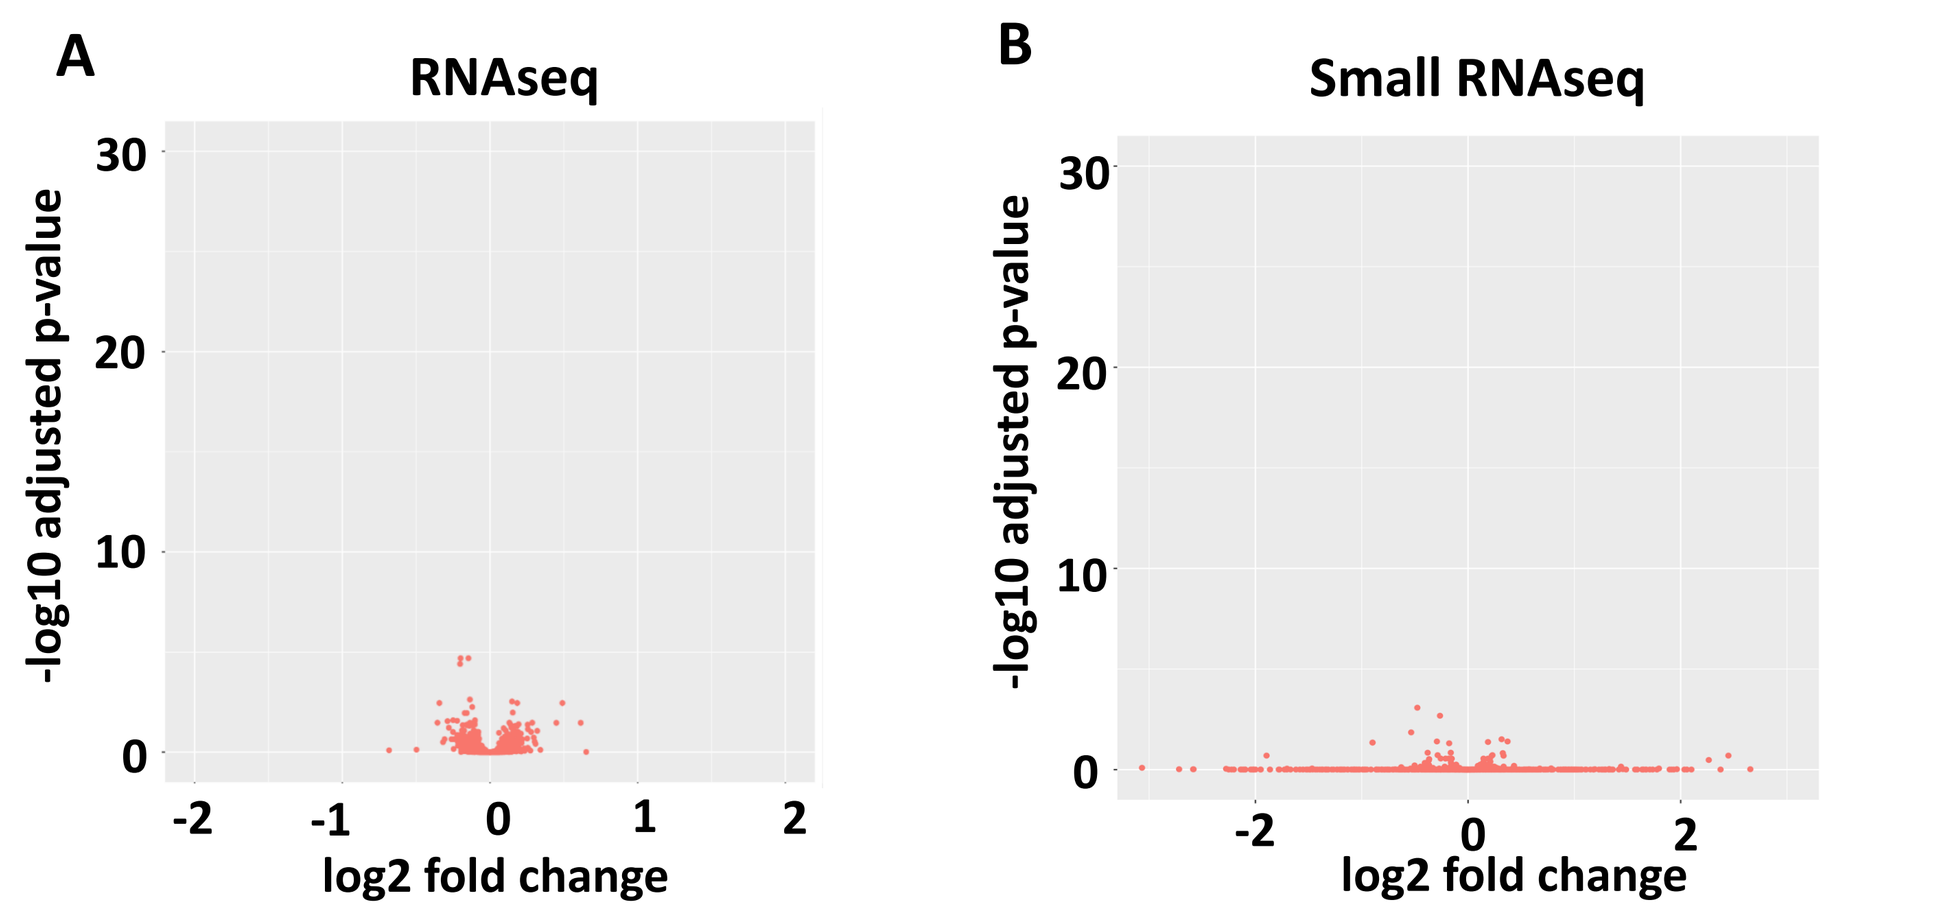
A
RNAseq
B
Small RNAseq
-log10 adjusted p-value
log2 fold change

Figure 3: KateTx FSHD lead candidate does not result in any significant off-target effects in human myotubes.
A) Volcano plot of differentially expressed genes in human myotubes transduced with KateTx FSHD lead candidate vs scrambled control based on RNAseq.
B) Volcano plot of differentially expressed endogenous miRNAs in human myotubes transduced with KateTx FSHD lead candidate vs scrambled control based on small RNAseq.

KateTx FSHD lead candidate protects muscles against DUX4 induced damage in a mouse model of FSHD

- Bitransgenic ACTA1-MCM;FLExDUX4 mice express DUX4 and DUX4 downstream genes following tamoxifen induction.
- Injection of ACTA1-MCM;FLExDUX4 mice with the lead candidate prior to tamoxifen induction results in reduced DUX4 downstream gene expression and improvement of muscle histology and function.
- Treatment of aged, non-induced ACTA1-MCM;FLExDUX4 mice that show leaky DUX4 expression in their muscles improves muscles histology and results in reduced DUX4 downstream gene expression.

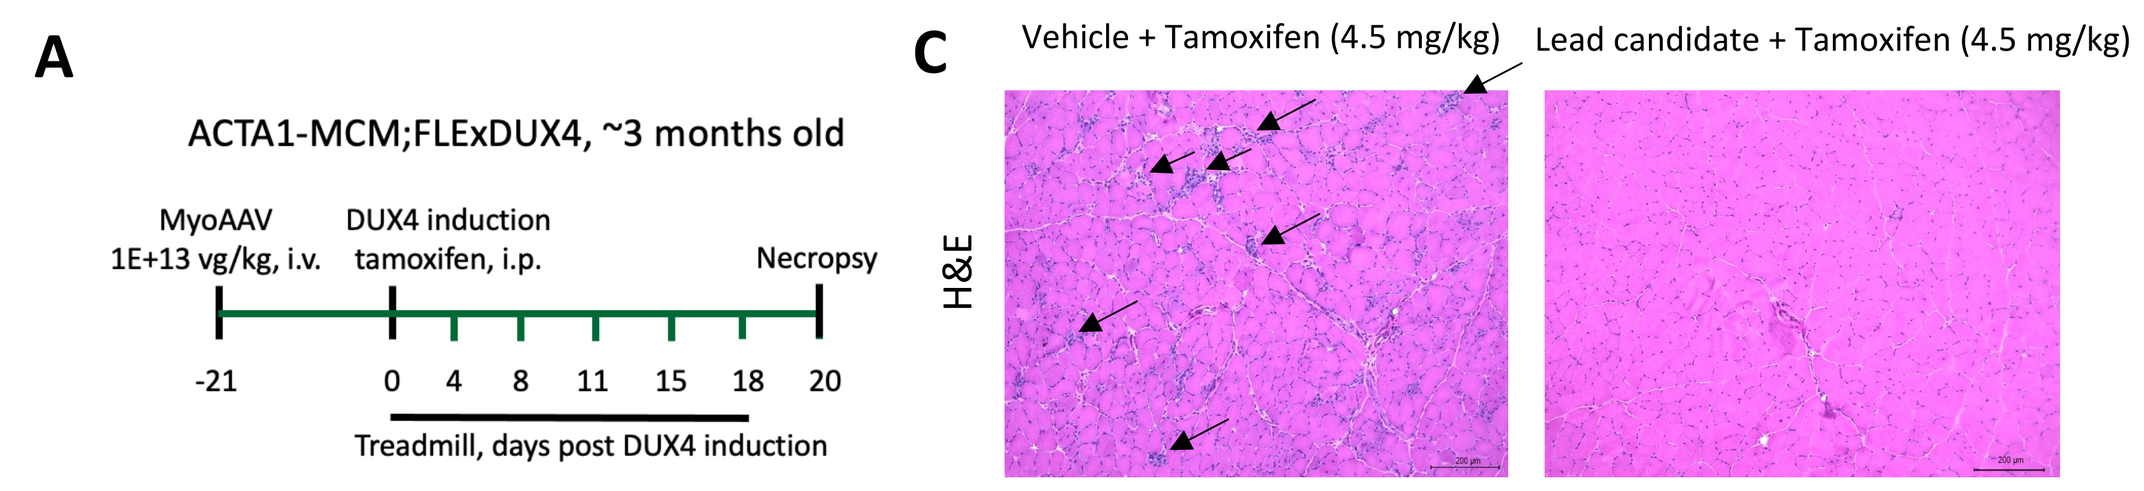
A
ACTA1-MCM;FLExDUX4, ~3 months old
MyoAAV 1E+13 vg/kg, i.v.
DUX4 induction tamoxifen, i.p.
Necropsy
Treadmill, days post DUX4 induction
C
Vehicle + Tamoxifen (4.5 mg/kg)
Lead candidate + Tamoxifen (4.5 mg/kg)
H&E

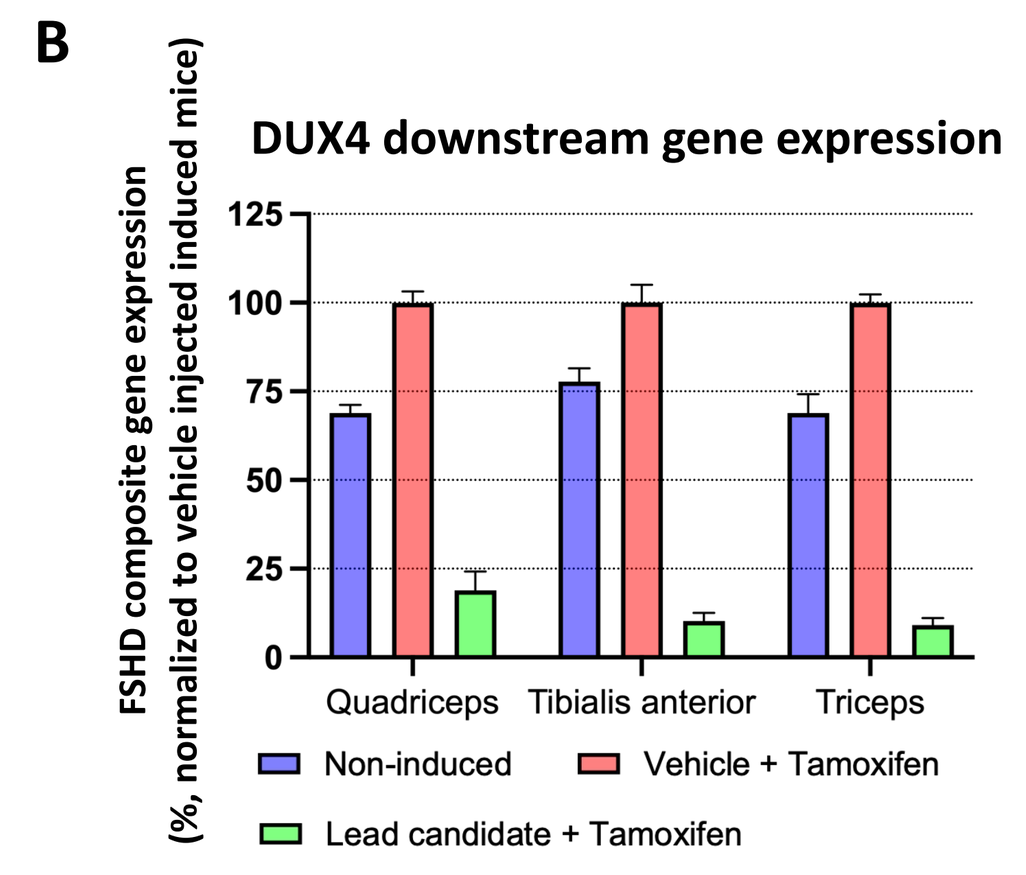
B
DUX4 downstream gene expression
FSHD composite gene expression (%, normalized to vehicle injected induced mice)
Quadriceps
Tibialis anterior
Triceps
Non-induced
Vehicle + Tamoxifen
Lead candidate + Tamoxifen

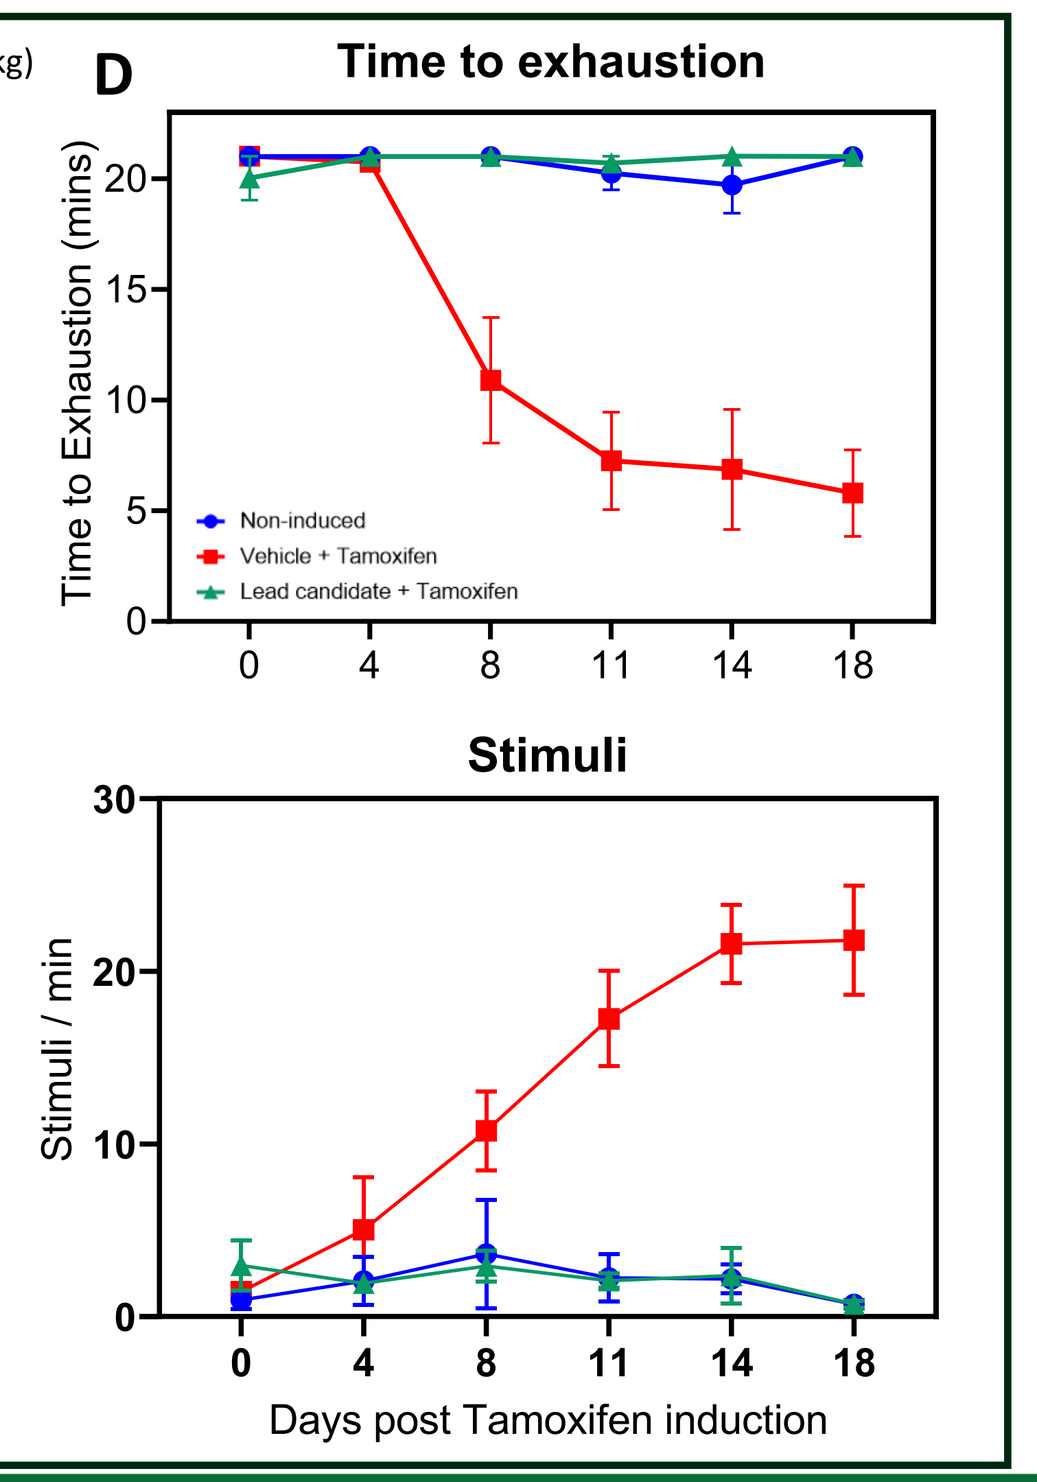
D
Time to exhaustion
Time to Exhaustion (mins)
Non-induced
Vehicle + Tamoxifen
Lead candidate + Tamoxifen
Stimuli
Stimuli / min
Days post Tamoxifen induction

Figure 4: KateTx FSHD lead candidate protects muscle fibers against DUX4 induced damage in a mouse model of FSHD.
A) Schematic of experimental design
B) DUX4 downstream gene expression is decreased in induced mice treated with the lead candidate. FSHD composite gene expression is the average of DUX4 downstream genes SLN, KIF4, WFDC3, and ENTPD3.
C) Muscle histology is improved in induced mice treated with the lead candidate compared to vehicle injected induced animals. Arrows show areas of active degeneration.
D) ACTA1-MCM;FLExDUX4 mice treated with the lead candidate run longer on the treadmill compared to vehicle injected induced animals.

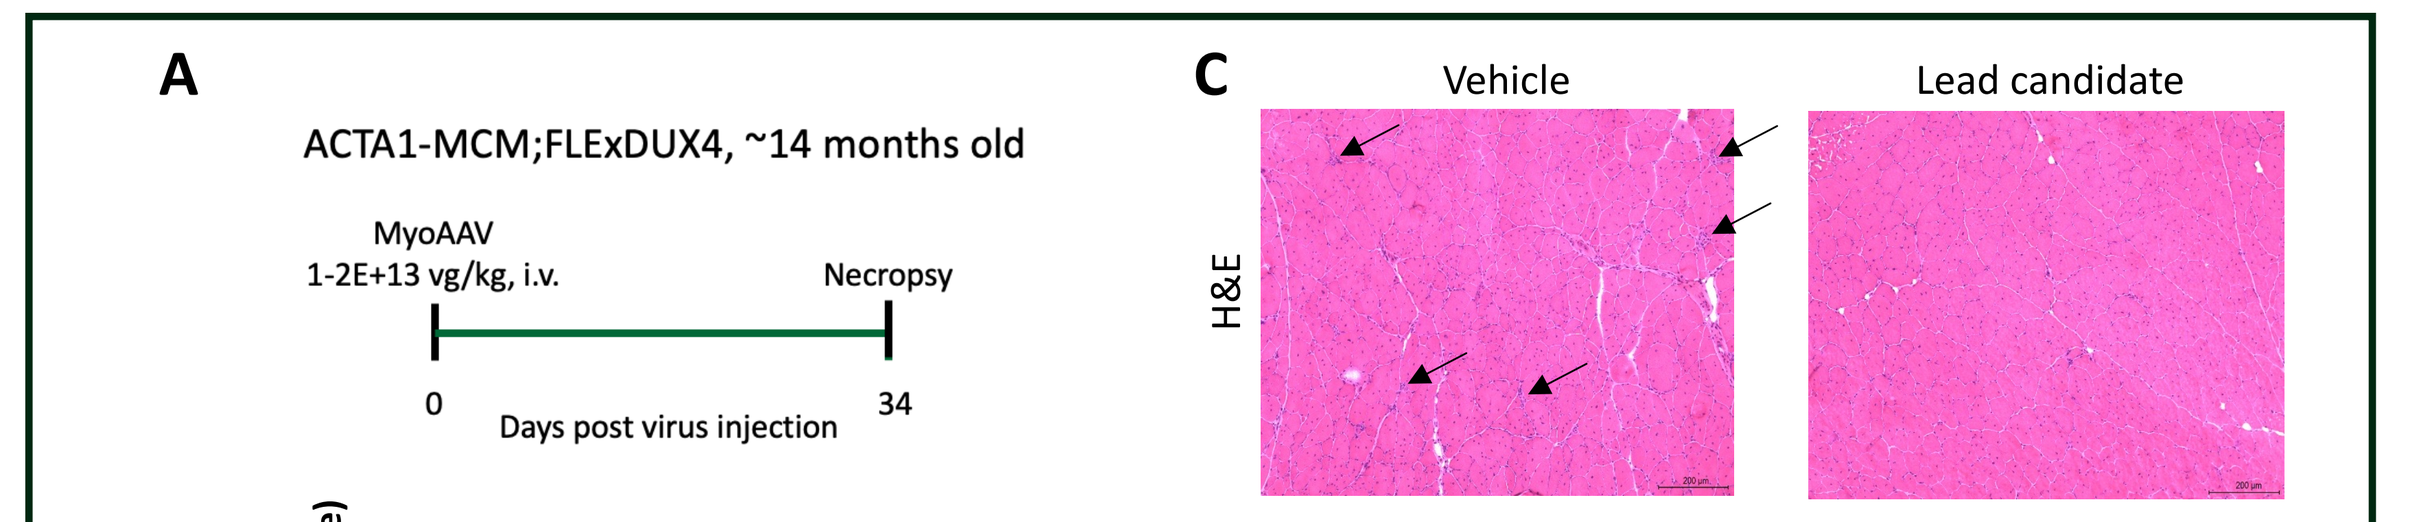
A
ACTA1-MCM;FLExDUX4, ~14 months old
MyoAAV 1-2E+13 vg/kg, i.v.
Necropsy
Days post virus injection
C
Vehicle
Lead candidate
H&E

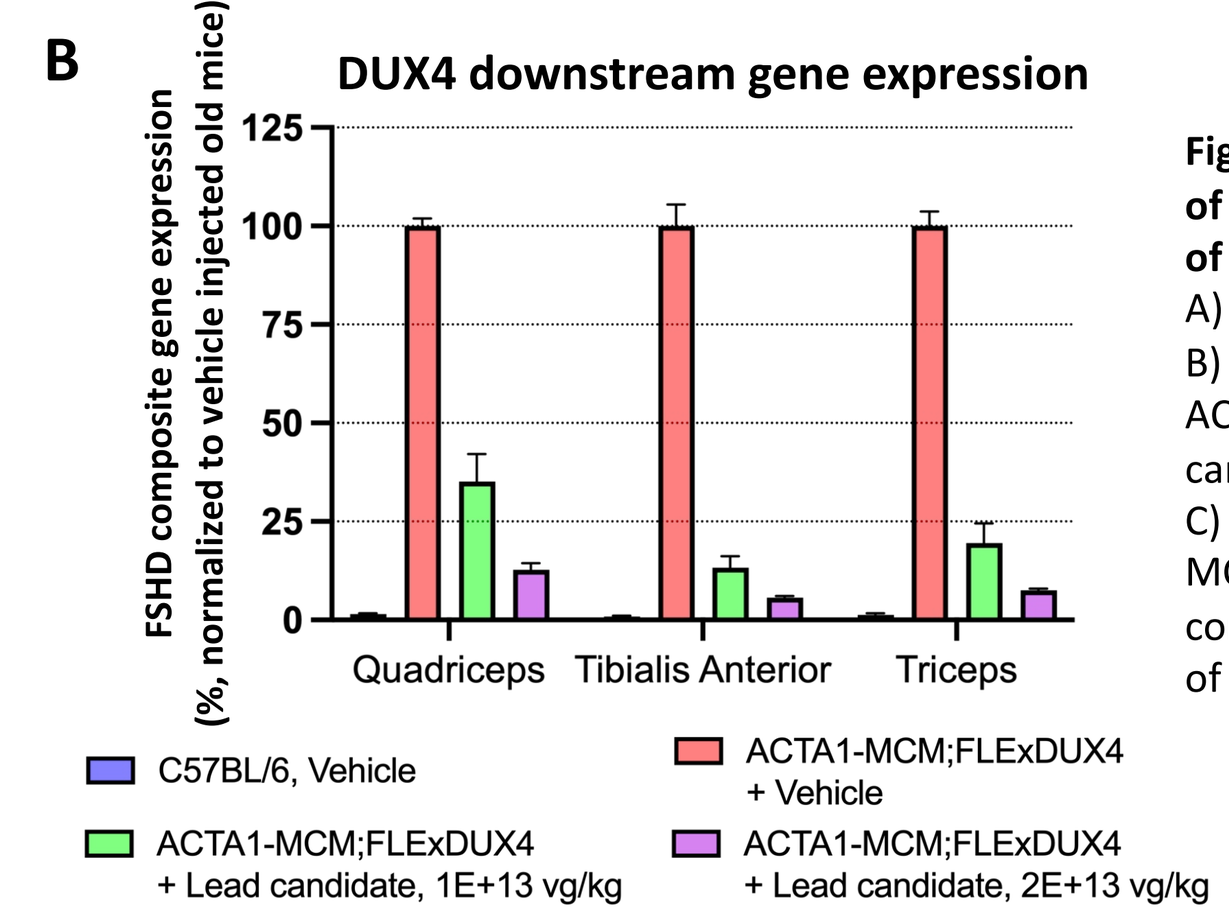
B
DUX4 downstream gene expression
FSHD composite gene expression (%, normalized to vehicle injected old mice)
Quadriceps
Tibialis Anterior
Triceps
C57BL/6, Vehicle
ACTA1-MCM;FLExDUX4 + Vehicle
ACTA1-MCM;FLExDUX4 + Lead candidate, 1E+13 vg/kg
ACTA1-MCM;FLExDUX4 + Lead candidate, 2E+13 vg/kg

Figure 5: KateTx FSHD lead candidate results in reduction of DUX4 downstream gene expression and improvement of muscle histology in old ACTA1-MCM; FLExDUX4 mice.
A) Schematic of the experiment.
B) DUX4 downstream gene expression is decreased in old ACTA1-MCM;FLExDUX4 mice treated with the lead candidate.
C) Muscle histology is improved in old ACTA1-MCM;FLExDUX4 mice treated with the lead candidate compared to vehicle injected animals. Arrows show areas of active degeneration.

Engineered muscle tropic liver de-targeted capsid used in KateTx FSHD lead candidate transduces all muscle fibers after systemic administration in NHPs

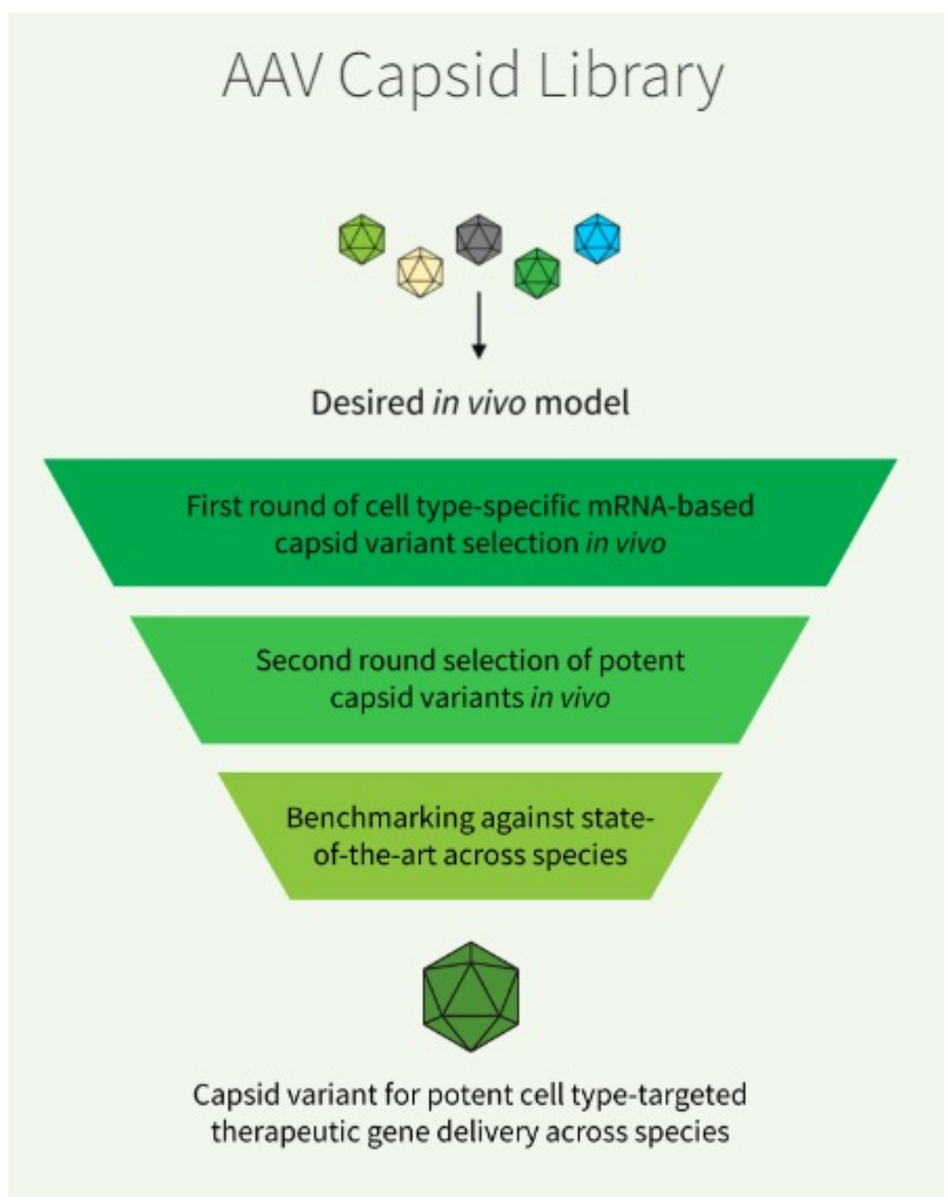
AAV Capsid Library
Desired in vivo model
First round of cell type-specific mRNA-based capsid variant selection in vivo
Second round selection of potent capsid variants in vivo
Benchmarking against state-of-the-art across species
Capsid variant for potent cell type-targeted therapeutic gene delivery across species

- Evolved a new class of muscle targeted and liver de-targeted MyoAAV-LD capsid variants using the DELIVER platform.
- Top MyoAAV-LD variants from directed evolution were benchmarked against naturally occurring and engineered AAV capsids in NHPs.
- MyoAAV-LD 6.1 was identified as the lead capsid and is used in the KateTx FSHD lead candidate.

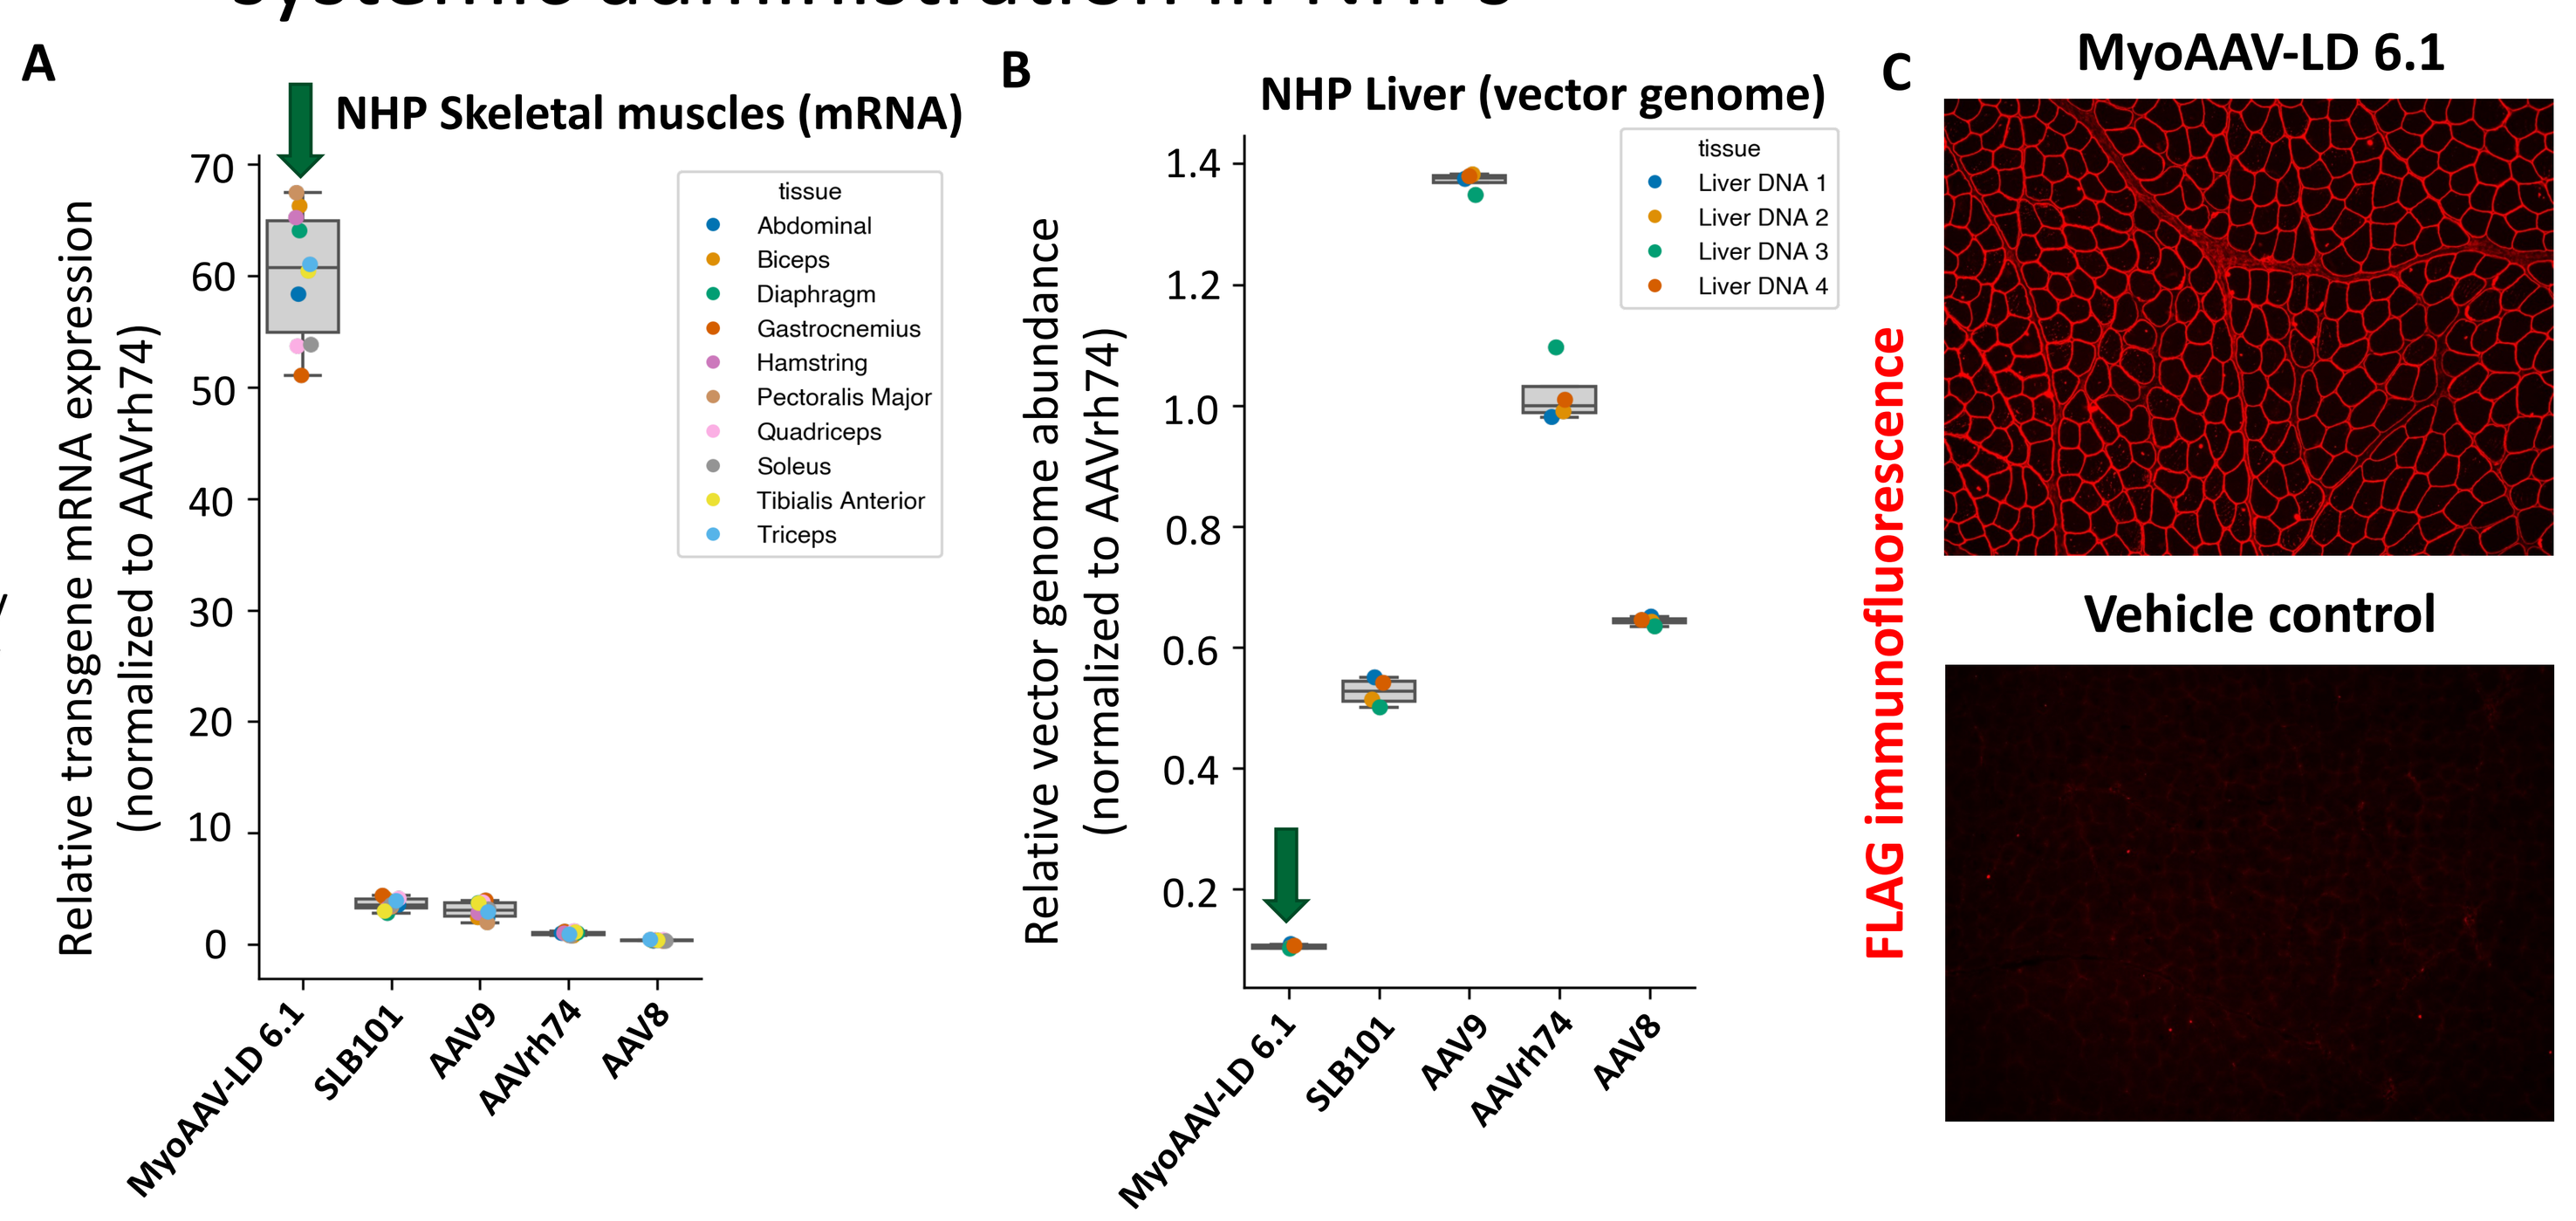
A
NHP Skeletal muscles (mRNA)
Relative transgene mRNA expression (normalized to AAVrh74)
tissue
Abdominal
Biceps
Diaphragm
Gastrocnemius
Hamstring
Pectoralis Major
Quadriceps
Soleus
Tibialis Anterior
Triceps
MyoAAV-LD 6.1
SLB101
AAV9
AAVrh74
AAV8
B
NHP Liver (vector genome)
Relative vector genome abundance (normalized to AAVrh74)
Liver DNA 1
Liver DNA 2
Liver DNA 3
Liver DNA 4
C
MyoAAV-LD 6.1
Vehicle control
FLAG immunofluorescence

Figure 6: MyoAAV-LD-6.1 transduces all muscle fibers after systemic administration in NHPs.
A) Transgene mRNA expression from MyoAAV-LD 6.1 compared to different naturally occurring and engineered capsids in NHP skeletal muscles. A uniquely barcoded transgene was packaged in each capsid and the pool of capsids was injected into NHPs. Abundance of transgene mRNA was measured by NGS. MyoAAV-LD-6.1 showed ~60 times higher transgene expression compared to AAVrh74.
B) MyoAAV-LD-6.1 showed ~10 times lower vector genome levels in the liver compared to AAVrh74.
C) FLAG immunofluorescence on Biceps Brachii from Cynomolgus macaques systemically injected with MyoAAV-LD-6.1–KEP1.1-microDystrophin-FLAG at 4E+13 vg/kg.

Conclusions

- High throughput tiling screen of artificial miRNAs targeting DUX4 identified the most potent miRNA targeting the transcript.
- MyoAAV-LD 6.1 used in KateTx FSHD lead candidate significantly outperforms competitor capsids in transducing NHP muscle and is de-targeted from the liver after systemic administration.
- KateTx FSHD lead candidate knocks down DUX4 and DUX4 downstream genes in FSHD patient myotubes, regardless of timing of transduction.
- KateTx FSHD lead candidate knocks down DUX4 downstream genes and improves functional phenotypes and histopathology in a severe mouse model of FSHD.
- KateTx FSHD lead candidate does not result in off-target effects on endogenous mRNA and miRNA expression in human myotubes.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com